Tego® Connectors Reduce Heparin Use Without Affecting Blood Flow Rate Compared to Traditional Central Venous Catheter Locks

C Farthing, BSN, RN, CNN;[1] M Krishnan, MD, MBA, MPH, FASN;[2] T Mayne, PhD;[2] S Collard, PharmD;[1] A Nissenson, MD, FACP[1]

[1]DaVita Inc, Denver, CO, USA; [2]DaVita Clinical Research, Minneapolis, MN, USA

Introduction

- Ten percent of patients with a long-term central venous catheter (CVC) for chronic hemodialysis (HD) will develop a catheter-related blood stream infection (CRBSI).[1–4]
- The Tego® Connector is a device developed to reduce catheter-related infections and clots associated with CVC without the use of heparin while maintaining blood flow rate (Figure 1).
- The Tego is a *neutral* displacement connector; when the blood tubing or a syringe is removed from the Tego connector there is minimal reflux of blood into the catheter lumen, reducing the need for a heparin or sodium citrate CVC lock.
- The Tego Connector is changed every 7 days and the CVC is locked with saline only following each treatment.
- ***We compared the efficacy and cost effectiveness of Tego Connectors and saline CVC locks to traditional heparin CVC locks, and compared both to the costs of rt-PA.***

Figure 1. Tego Connector

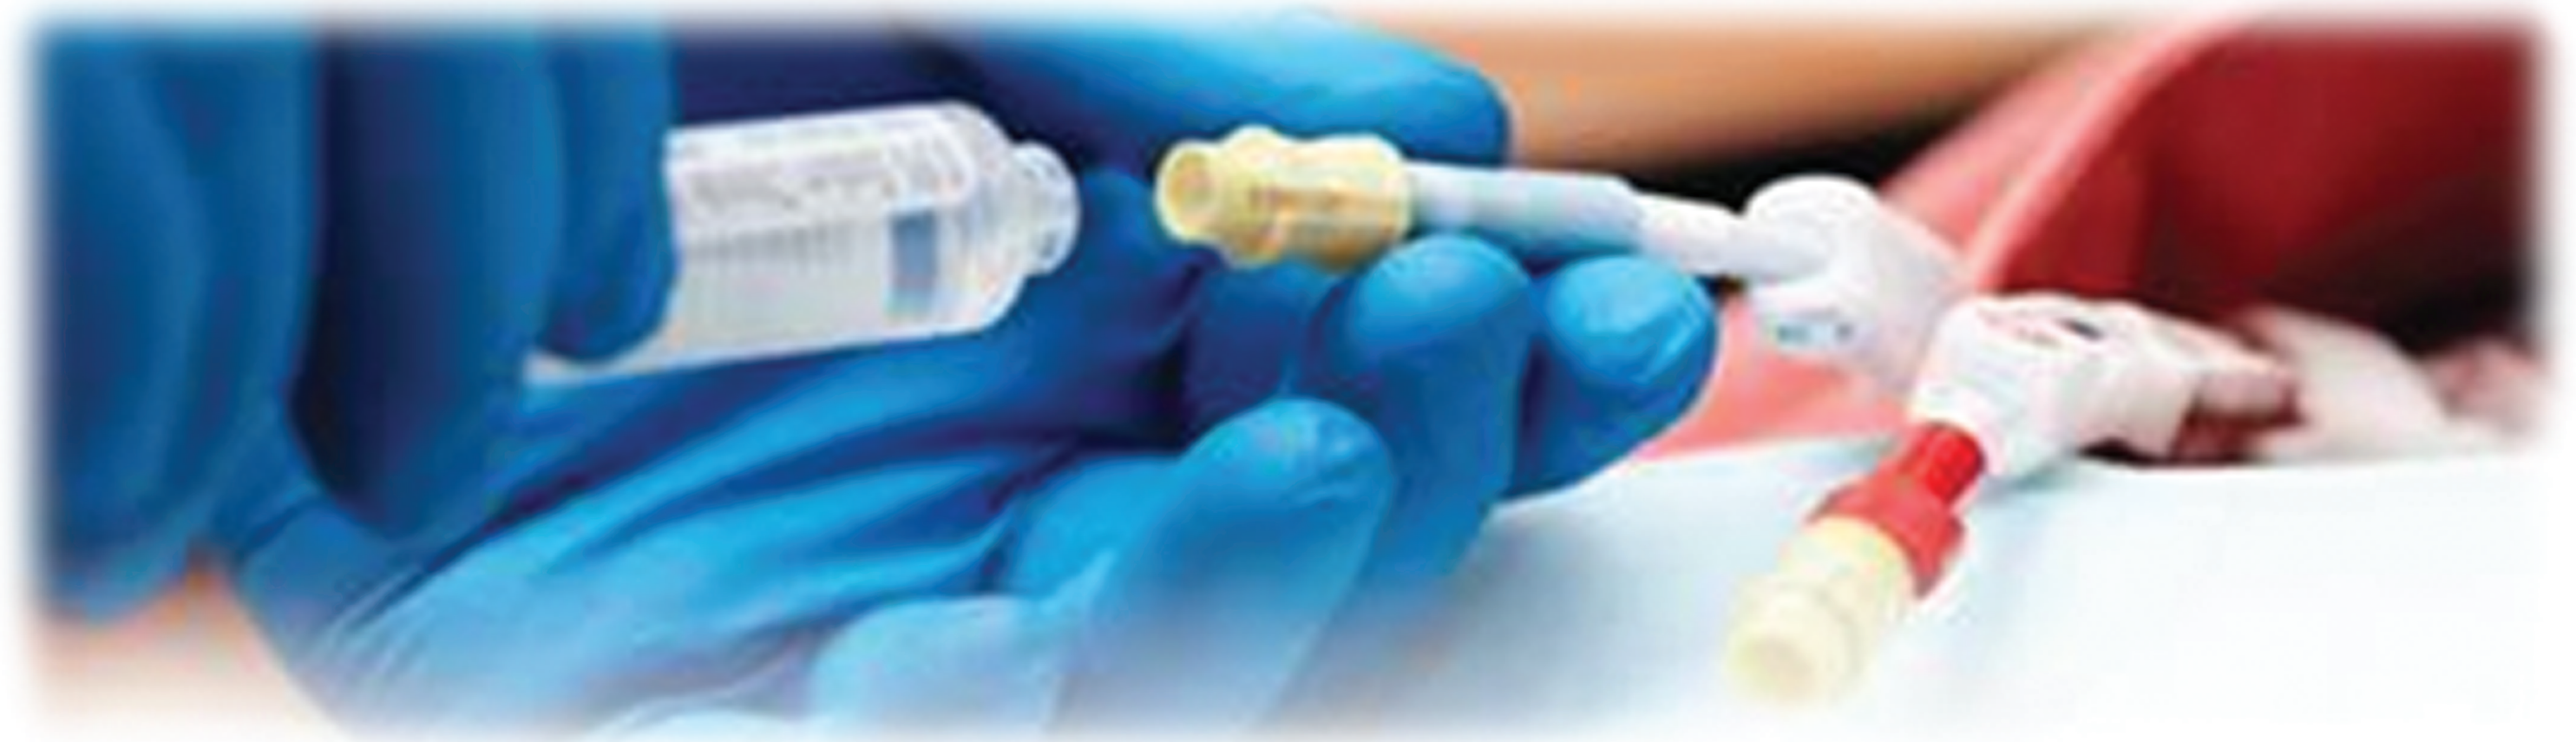

Methods

- In this retrospective analysis, we compared conversion from traditional CVC locks to Tego Connectors. The pre-period was defined as 90 days before the conversion and the post-period was defined as 90 days after Tego Connector conversion (Table 1).
- For the efficacy analysis, we assessed monthly blood flow rate, heparin use, and blood culture results.
- For the cost analysis, the cost of heparin, connectors, syringes, activase, sodium citrate, and equipment for both CVC locks and Tego Connectors were compared to the cost of recombinant tissue plasminogen activator (rt-PA) as of July 2010.

Results

Table 1. Demographics

Mean ± SD	Patients
N	2,737
Mean age ± SD (yr)	62.31 ± 16.24
% Male	46.7%
Race and Ethnicity	
% African American	20.9%
% Hispanic	27.5%
% Asian, P. Islander	9.0%
% Native American	0.9%
% Caucasian	38.9%
% Other	2.8%
% with Diabetes	49.3%
Mean vintage ± SD (yr)	2.93 ± 3.67
Vintage (yr) min–max	0.25 - 36.72
BMI ± SD	26.86 ± 7.19

Figure 2. Relative Mean 6-Month Cost of Locks per CVC Patient

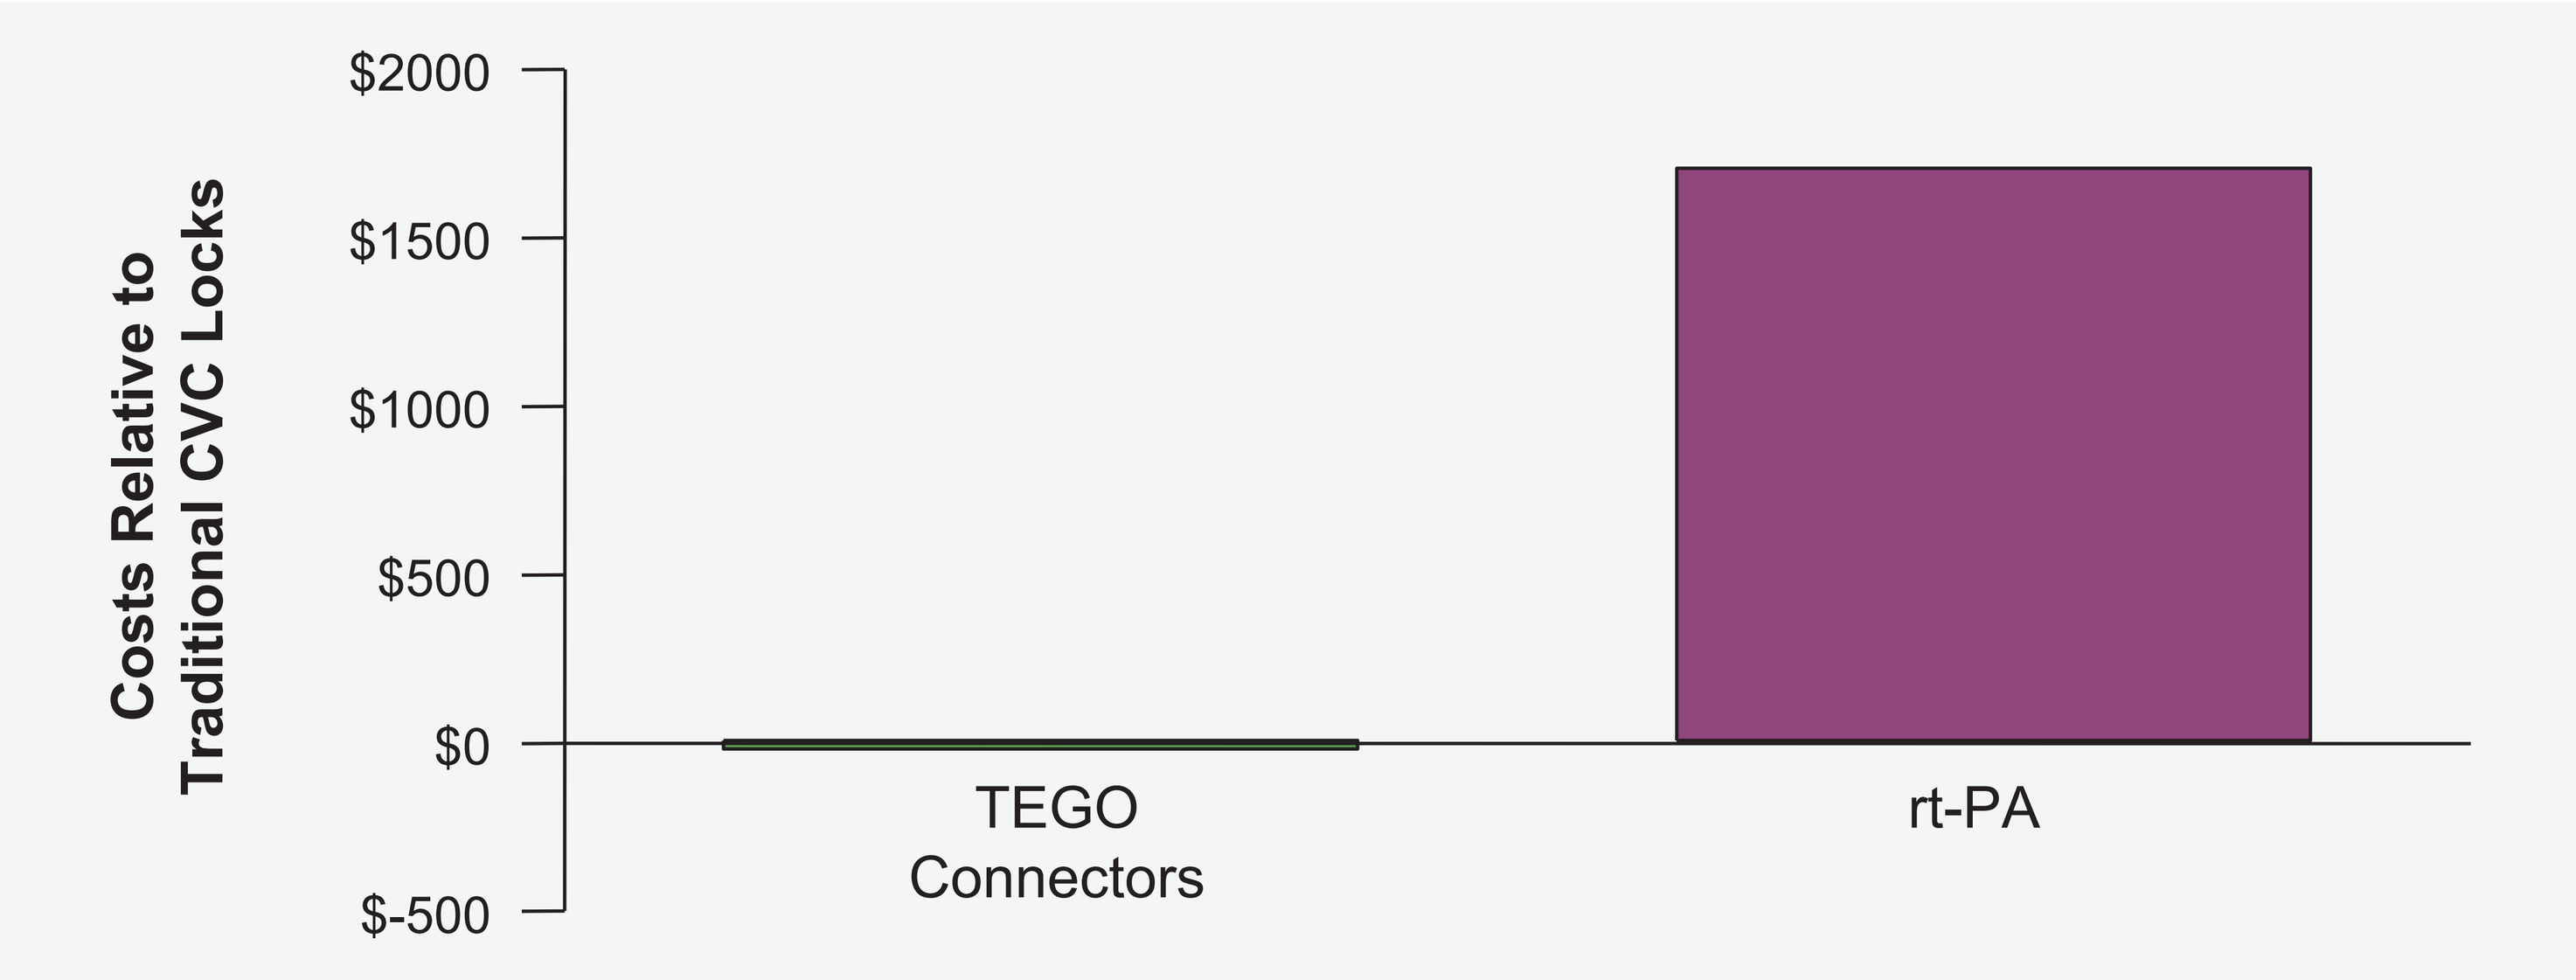
Costs Relative to Traditional CVC Locks
$2000
$1500
$1000
$500
$0
$-500
TEGO Connectors
rt-PA

- Blood flow rate and the rate of positive blood cultures remained unchanged over the course of the evaluation period (Table 2).
- Total heparin use decreased nearly 2000 units 3 months after Tego conversion (Table 2).
- The mean 6-month cost was $16.38 lower for Tego Connectors/CVC patient compared to traditional locks + caps + heparin + syringes/CVC patient. Both are significantly less than the rt-PA cost/patient (Figure 2).

Table 2. Blood Flow Rate, Heparin Use, and Positive Blood Cultures (Before and After Conversion to Tego Connectors)

	Days Prior to Conversion			Days After Conversion		
	90–61	60–31	30–01	01–30	31–60	61–90
# Facilities	225	230	234	234	233	232
# Patients	2,040	2,302	2,656	2,656	2,447	2,256
Blood Flow Rate (mL/min); mean ± SD	348.3 ± 41.5	348.2 ± 42.6	346.7 ± 40.3	345.4 ± 40.7	343.1 ± 41.4	341.4 ± 41.1
Run Time (minutes); mean ± SD	208.6 ± 30.7	207.5 ± 30.1	207.2 ± 28.8	207.4 ± 29.5	206.9 ± 28.6	208.1 ± 29.8
Kt/V; mean ± SD	1.5 ± 0.4	1.5 ± 0.4	1.5 ± 0.4	1.6 ± 0.4	1.6 ± 0.4	1.6 ± 0.4
Total Heparin Units / Treatment	6,177	6,782	7,799	6,692	4,933	4,240
Total Activase (mg) / Treatment	0.06	0.06	0.07	0.06	0.05	0.05
Positive Cultures per 1,000 Patient Days (Number)	1.04	0.88	1.04	0.93	0.93	0.87
Positive Blood Cultures (%)	16.3%	15.7%	17.9%	17.5%	17.1%	14.9%

Conclusions

- Use of Tego Connectors decreased heparin use resulting in a lower cost per month.
- Blood flow rate and the rate of positive blood cultures were essentially unchanged after conversion to Tego Connectors.
- Costs of both Tego and saline CVC locks were significantly lower than use of rt-PA.
- Tego Connectors are a viable and cost-effective alternative to traditional locks.

References

1. Nielsen, J, Ladefoged SD, Kolmos HJ. Dialysis Catheter-Related Septicaemia. Focus on S. Aureus Septicaemia. *Nephr Dial Transplant.* 1998;13:2847.
2. Marr KA, Sexton DJ, Conlon PJ, et al. Cathéter-Related Bacteremia and Outcome of Attempted Salvage in Patients Undergoing Hemodialysis. *Ann Inter Med.* 1997;12,275–280.
3. Beathard G. Management of Bacteremia Associated with Tunneled-Cuffed Hemodialysis Catheters. *J Am Soc Nephrol.* 1999;10:1045.
4. Saad TF. Bacteremia Associated with Tunneled, Cuffed Hemodialysis Catheter. *Am J Kidney Dis.* 1999;34:1114–24.

Acknowledgements

Our sincere appreciation to the teammates in more than 1600 DaVita clinics who work every day to take care of patients but also to ensure the extensive data collection on which our work is based. We thank DaVita Clinical Research® (DCR®), and specifically acknowledge Lesley Smith, MA and Karen Spach, PhD of DCR for editorial contributions in preparing this poster. DCR is committed to advancing the knowledge and practice of kidney care.

*Correspondence: mahesh.krishnan@davita.com

Poster available at www.davitaclinicalresearch.com/directory.asp

American Nephrology Nurses' Association 43rd National Symposium, April 29–May 2, 2012; Orlando, FL.

©2012 DaVita Inc. All rights reserved. Proprietary. May not be copied, reprinted or distributed without the permission of DaVita Inc.